A systematic review of economic evaluations in non-insulin antidiabetic treatments for patients with type 2 Diabetes Mellitus

Néboa Zozaya[1], Margarita Capel[2], Susana Simón[2], Alfonso Soto-González[3], Álvaro Hidalgo-Vega[4]

[1]Department of Health Economics. Weber, Madrid, Spain. [2]Astrazéneca, Madrid, Spain. [3]Department of Endocrinology and Nutrition. Gerencia de Gestión Integrada de A Coruña. [4]Fundación Weber, Madrid, Spain. / Universidad de Castilla-La Mancha. Campus de Toledo, Spain.

PDB83

Context

Diabetes mellitus type 2 (DM2) is one of the chronic diseases with the greatest impact on public health in developed countries, due to its high prevalence and associated morbidity and mortality. It is estimated that diabetes accounts for 11.6% of total healthcare expenditure worldwide[1].

Regarding the treatment of DM2, when glycemic control is inadequate in a monotherapy regimen, in general, a dual or triple therapy would be used, combining the pharmacological treatment of non-insulin antidiabetics (NIADs) or insulin[2,3]. The introduction of new NIADs has allowed the available therapeutic arsenal to be expanded, but at the same time it has increased the complexity of choice of treatment. In a context of limited budgetary resources, prioritising the use of efficient healthcare interventions is essential for the rational use of those resources and, therefore, for the sustainability of the healthcare system. The economic evaluation is a fundamental tool for making rational decisions. Thus, with the appearance of new treatments, the economic evaluations must also be updated.

The main objective of this work was to evaluate the efficiency of NIADs in the treatment of DM2, through a systematic review of the published economic evaluations in NIADs.

Methods

- A systematic review of the literature was carried out, following the CRD's guidance for undertaking reviews in health care of the University of York.
- The search strategy took into account free text and MESH terms.
- The searching engines used were Medline, IBECS, Doyma and SciELO databases.
- The inclusion criteria are shown on Table 1.

Table 1. Inclusion criteria for the systematic review

Full economic evaluations directly related to:	And which include:
NIADs for the treatment of DM2: • Metformin • Glinides (repaglinide) • Glitazone (pioglitazone) • Sulfonylureas (glibenclamide, glipizide, glimepiride, gliclazide) • iDPP-4 (sitagliptin, vildagliptin, saxagliptin, alogliptin, linagliptin) • aGLP-1 (exenatide, liraglutide, dulaglutide, albiglutide, lixisenatide) • iSLGT-2 (dapagliflozin, empagliflozin,canagliflozin)	• A quantifiable measurement of clinical effectiveness measured in terms of QALY of the alternatives compared. • A measurement of the cost of the alternatives compared. • Incremental cost-utility ratio, or data to calculate it.
Limited to:	
Patients: Adults in treatment of DM2 after the failure of the 1st line of pharmacological treatment. Publication in scientific journals. Full text language: Spanish, English. Publication dates: 2010-2017	Countries: Europe, United States, Canada. Comparators: Placebo, insulins or other oral/subcutaneous NIADs (in monotherapy or in combination).

DM2= Diabetes Mellitus 2, QALY= Quality-adjusted life years, NIADs= Non-insulin antidiabetics

- Two reviewers independently selected the studies and extracted data following the PRISMA guidelines.
- The results were updated to euros of 2017. A maximum cost-effectiveness threshold of €25,000 per quality-adjusted life year (QALY) was considered.
- An evaluation of the quality of the studies was carried out, applying the Drummond checklist.

Results

- The search identified a total of 601 studies. After eliminating the duplicates, the titles and abstracts of the 553 resulting articles were reviewed, from which 155 studies of potential interest were selected. After reviewing their full-texts, 98 papers were excluded, for different reasons.
- The review included a total of 57 studies, in which 134 comparisons were made between NIADs.
- We analyzed a total of 223 comparisons (Figure 1), since comparisons are bidirectional.
- The aGLP-1 liraglutide was the NIAD most frequently evaluated (48 times), followed by the aGLP-1 exenatide (38 times) and the iDPP-4 sitagliptin (21 times).
- Insulins,, sitagliptin and glipizide were the most common comparators.
- No study included gliclazide, linagliptin or empagliflozin.

Figure 1. Drugs participating in the comparisons drawn from the 57 studies

Group	Drug	Treatment	Comparator	Total
BIGUANIDES: 4	Metformin	0	4	4
GLITAZONES: 20	Unspecified	0	4	4
	Pioglitazone	5	7	12
	Rosiglitazone	0	4	4
SULFUNYLUREAS: 31	Unspecified	4	4	8
	Glibenclamide	1	4	5
	Glimepiride	0	5	5
	Glipizide	0	13	13
	Gliclazide	-	-	0
GLINIDES: 2	Repaglinide	2	0	2
iDPP-4: 44	Unspecified	4	4	8
	Saxagliptin	6	0	6
	Sitagliptin	5	16	21
	Vildagliptin	6	1	7
	Alogliptin	2	0	2
	Linagliptin	-	-	0
aGLP-1: 102	Unspecified	2	0	2
	Albiglutide	3	0	3
	Dulaglutide	2	1	3
	Exenatide	27	11	38
	Liraglutide*	44	4	48
	Lixisenatide	3	5	8
iSLGT-2: 20	Canagliflozin	2	0	2
	Dapagliflozin	14	4	18
	Empagliflozin	-	-	0
OTHERS not included: 42	Insulin/ acarbose/placebo	2	39	41
	Total	134	130	264
	Included NIADs	132	91	223

*Co-formulation liraglutide-insulin degludec

- Regarding the 57 studies included in the review, the following aspects should be noted: 77% of the studies were carried out in Europe; the perspective most frequently used was that of the healthcare financer (93%); the most frequently time horizon was 40 years (37%), the most used simulation model were the CORE Diabetes Model (44%) and the Cardiff Diabetes Model (28%); the most common discount rate was 3% and 3.5%, in 40% and 32% of the studies, respectively; most of the studies performed a sensitivity analysis on the results (91%).
- The quality of the economic evaluations was deemed acceptable.

Figure 2. Cost-effectiveness results found in the systematic review, by NIAD group

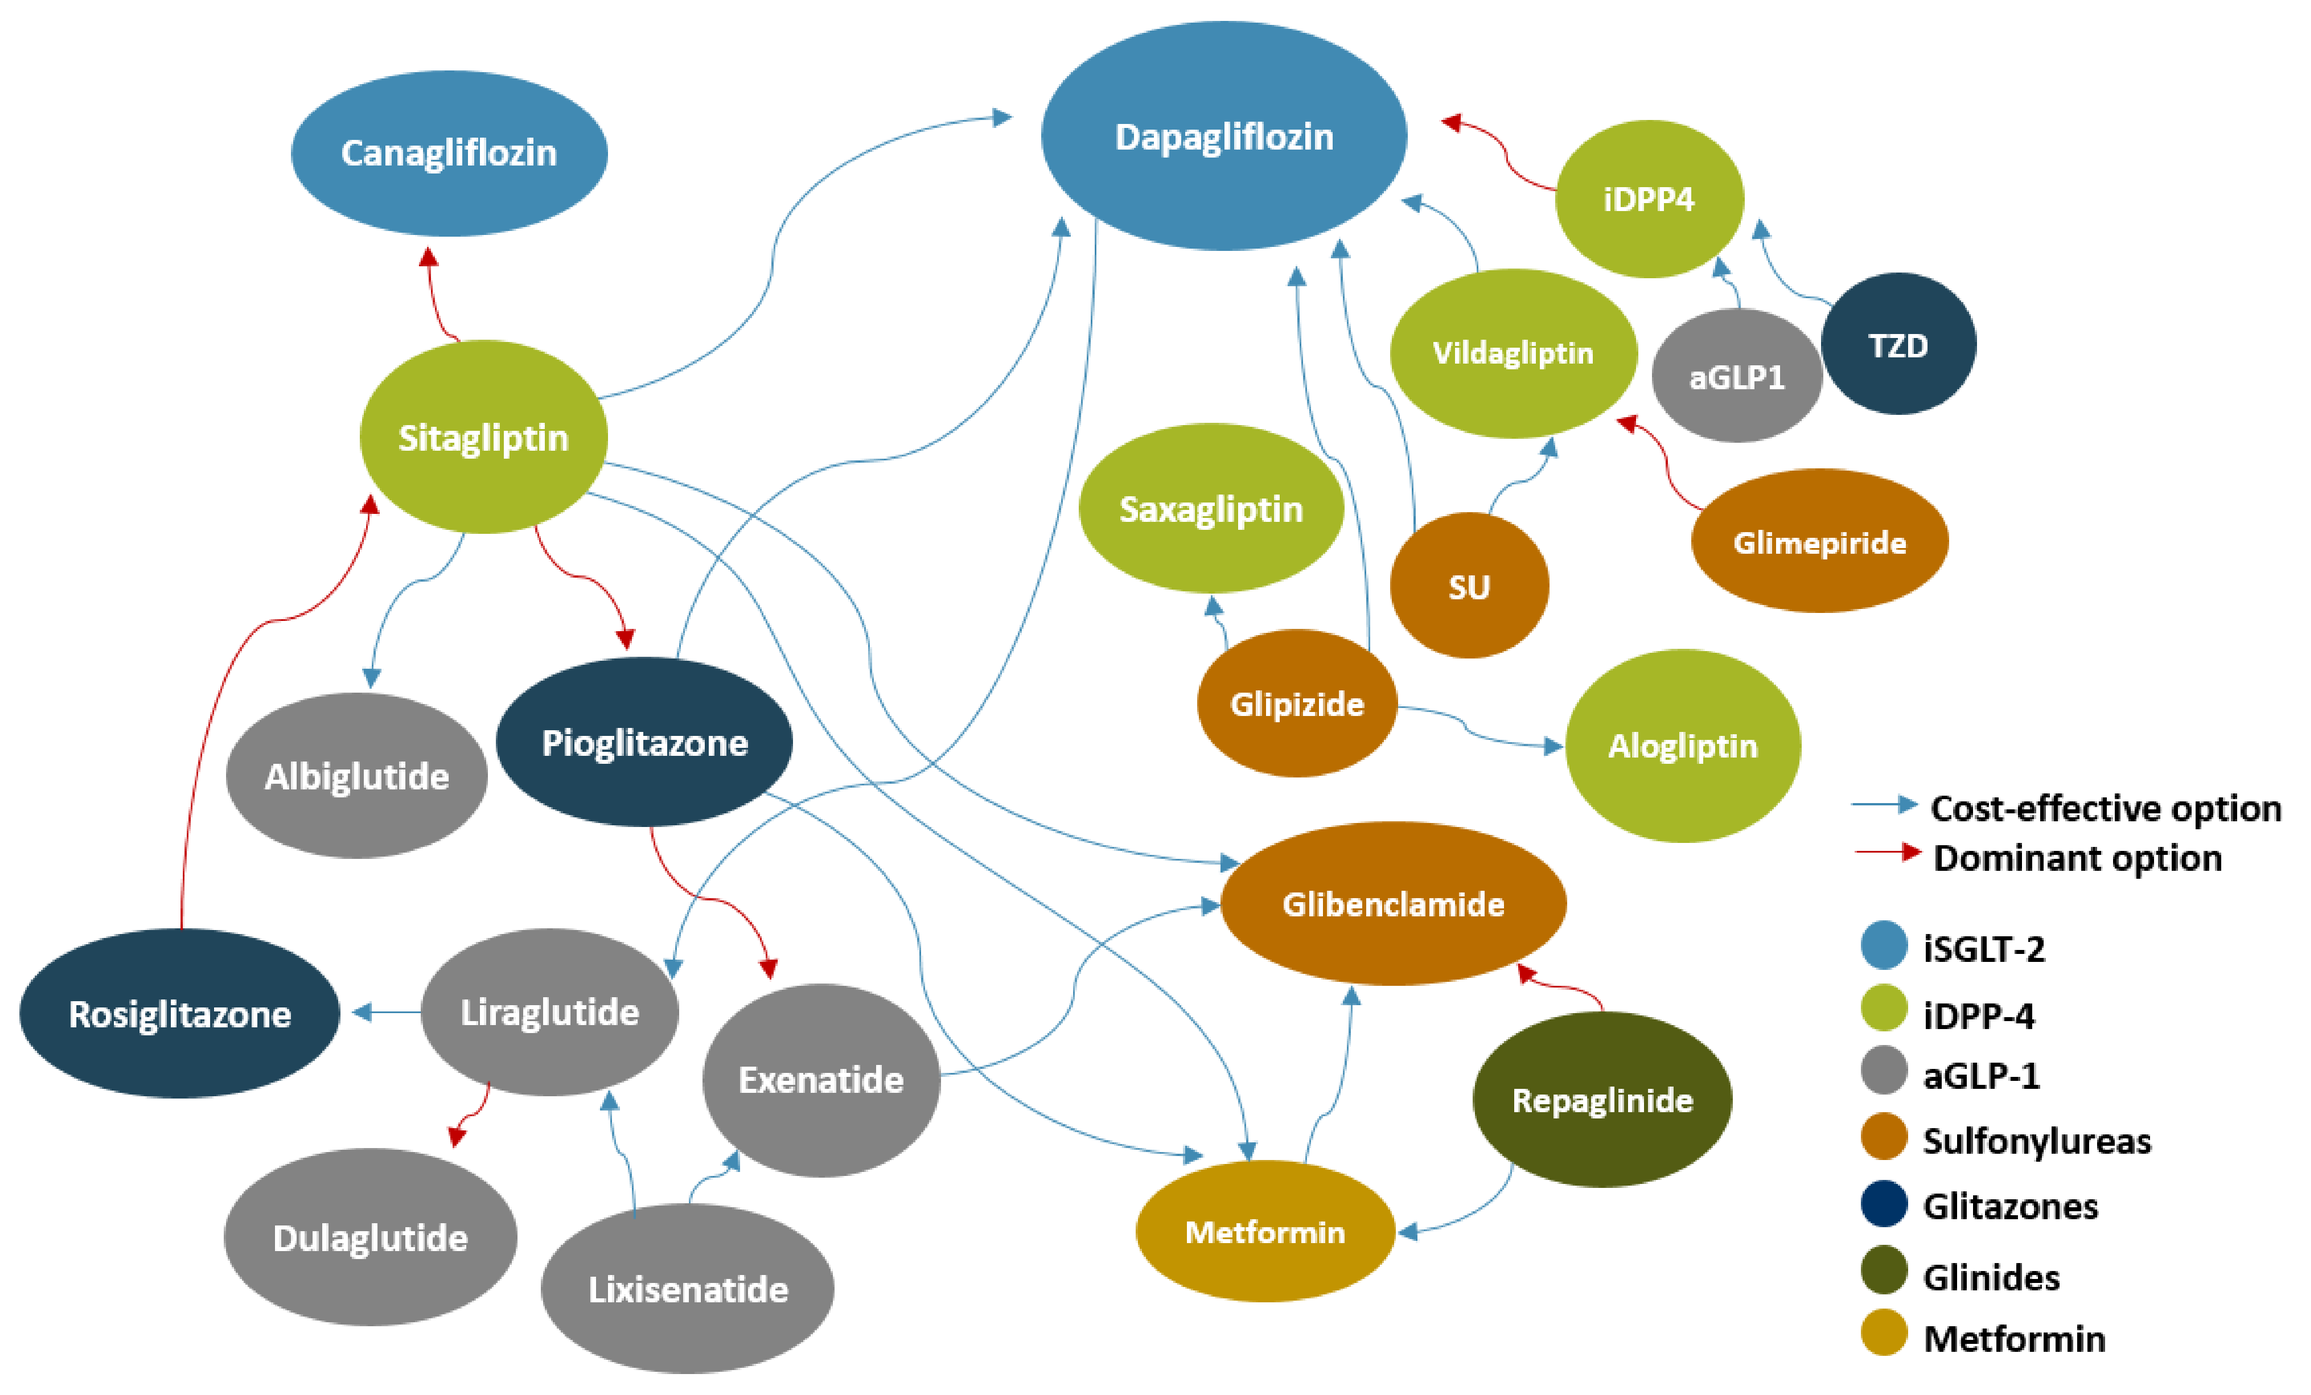
Canagliflozin
Dapagliflozin
iDPP4
Vildagliptin
aGLP1
TZD
Sitagliptin
Saxagliptin
SU
Glimepiride
Glipizide
Alogliptin
Albiglutide
Pioglitazone
Glibenclamide
Rosiglitazone
Liraglutide
Exenatide
Repaglinide
Dulaglutide
Lixisenatide
Metformin
Cost-effective option
Dominant option
iSGLT-2
iDPP-4
aGLP-1
Sulfonylureas
Glitazones
Glinides
Metformin

Note: The arrow points to the NIAD that is cost-effective or dominant

- Figure 2 summarizes the efficiency results obtained in this review.
- ISGLT-2 (dapagliflozin and canagliflozin) were preferable, in terms of cost-effectiveness, to iDPP-4, sulfonylureas and pioglitazone. However, the same cannot be said about their superiority over aGLP-1. Dapagliflozin was a more efficient option than iDPP-4 and was also cost-effective versus sulfonylureas. Canagliflozin was evaluated versus sitagliptin, resulting a dominant option.
- No conclusive results were obtained for iDPP-4, except when compared with iSGLT-2.
- The results were not conclusive for aGLP-1.
- It was not possible, either, to establish a clear option for sulfonylureas based on their efficiency, with the exception of dapagliflozin, which was more cost-effective than sulfonylureas.

Conclusions

- The only NIADs group for which conclusive favorable results were found in terms of efficiency are the most recently marketed, namely, the iSGLT-2 versus iDPP-4, sulfonylureas and glitazones.
- The results are valid under an acceptability threshold of €25,000/QALY.
- The heterogeneity of the studies' methodologies and results hindered our ability to determine under what specific clinical assumptions some NIADs would be more cost-effective than others.
- Economic evaluations of healthcare should be used as part of the decision-making process, so multifactorial therapeutic management strategies should be established based on the patients' clinical characteristics and preferences as principal criteria.

References

1) International Diabetes Federation. IDF Diabetes Atlas. Seventh Edition, 2015; 2. Algoritmo de tratamiento de la DM2, de la redGDPS.. Actualizado: 25/07/2017 .Access : http://www.redgdps.org/publicaciones-redgdps/ 3.Documento de abordaje integral de la diabetes tipo 2, Grupo de trabajo de Diabetes de la SEEN [2018].Access: http://www.seen.es

Acknowledgements
The authors want to thank Jhon Galindo (Weber Consultant) for the technical help performed in this study.

Presented at ISPOR Europe 2018, 10-14 November, 2018. Barcelona, Spain